Neurobiological Correlates of Age-related Auditory Verbal Working Memory Decline

LABORATOIRE DES NEUROSCIENCES DE LA PAROLE ET DE L'AUDITION
SPEECH AND HEARING NEUROSCIENCE LABORATORY

Maxime Perron[1], Julie Poulin[1], Isabelle Deschamps[1], Pascale Tremblay[1]

[1]Université Laval, Département de réadaptation, Centre de recherche CERVO

UNIVERSITÉ LAVAL
CENTRE DE RECHERCHE CERVO BRAIN RESEARCH CENTRE

Introduction

- Auditory verbal working memory (avWM) plays an important role in speech processing, allowing us to maintain and manipulate information in memory.

- Normal aging is associated with a decline in brain structure and function, which is associated with a decline in avWM [1].

- According to the Hemispheric Assymmetry Reduction in Older adults (HAROLD) model [2], prefrontal areas activation tends to be less lateralized in older adults than in younger adults during cognition tasks.

- The objective of this study is to investigate, using surface-based morphometry, the relationship between brain aging and avWM.

- Our hypothesis is that structural decline in the left lateralised cortical network supporting avWM (IFG, MFG, SMG, pSTG) [3,4], combined with an opposite right hemisphere structural decline, will be associated with a decline in avWM.

Methods

Study 1 (behavioural)

- 72 healthy participants aged 20 - 98 y (51.6 ± 20.3 y, 38 W).
- Audiological testing : pure tone average (PTA) for each ear .
- MoCA [5] (mean = 27.6 ± 1.94, range 23 to 30).
- avWM task : the Running Span test [6], requires participants to recall the last 3 - 6 digits (Span3 to Span6) from lists of different lengths.
- Dependent variables : Reaction Time (RT) and Accuracy (Acc).

Study 2 (MRI)

- 30 adults from Study 1 aged 21 - 86 y (52.6 ± 19.5 y, 16 W) (Objective: 40)
- Philips 3.0 T Achieva TX with a MPRAGE T1 sequence (1 mm3).
- MRI data processing with Freesurfer 6 (Figure 1) with the Destrieux 2009 parcellation [7].
- Surface, Volume and Thickness were extracted for each region of interest.

Figure 1 : MRI preprocessing

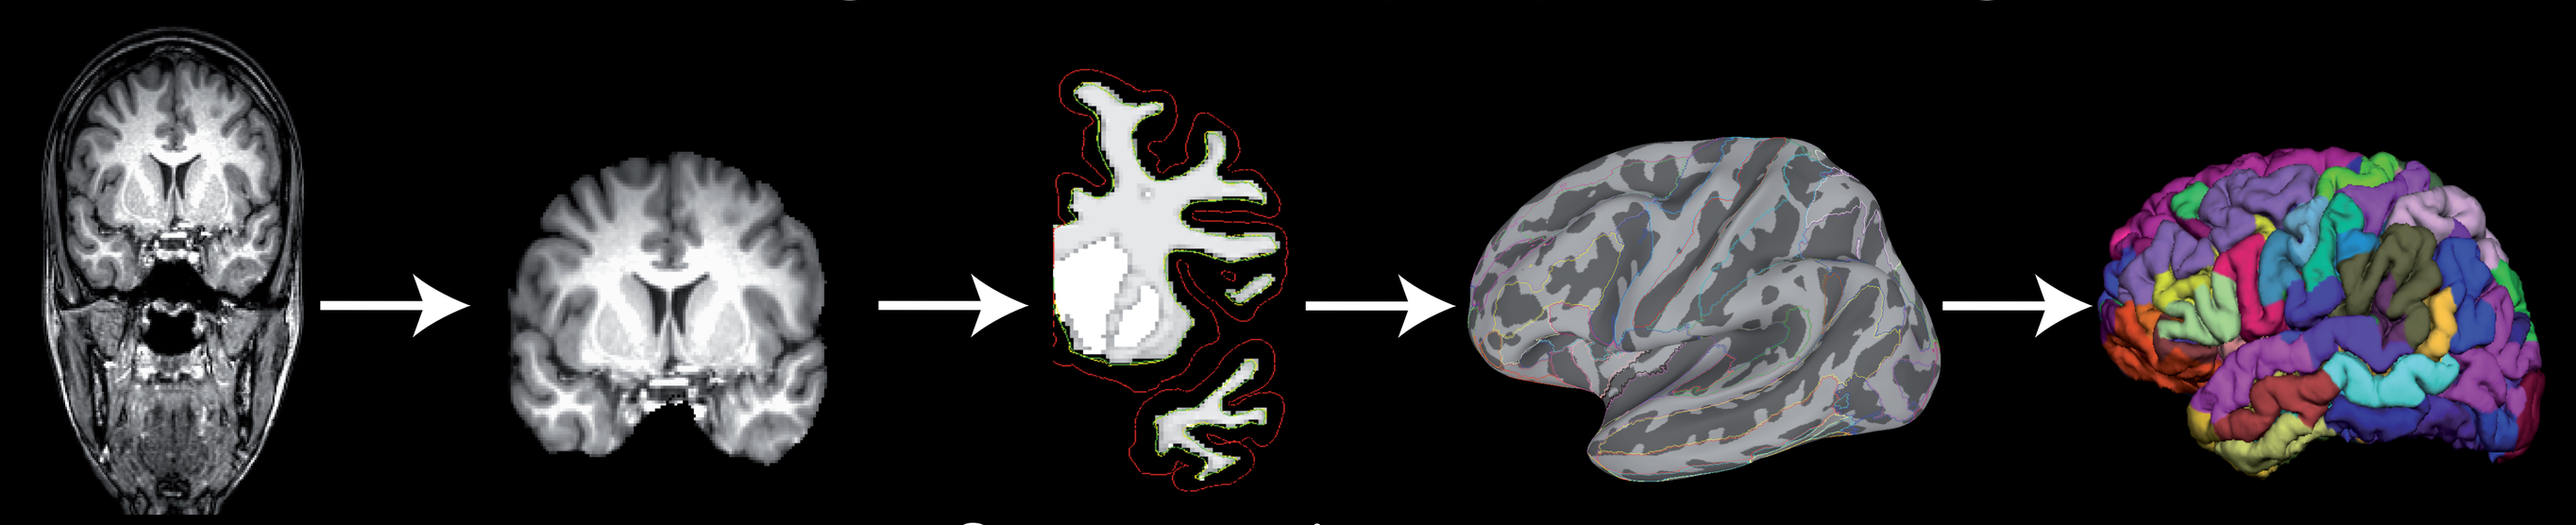
T1
Skullstrip
Segmentation GM/WM
Surfaces
Parcellation

Analysis and preliminary results

Study 1 (behavioural)

- Outliers were removed.

- Linear Mixed Model (LMM) analysis, separately for each DV, with Span (3, 4, 5, 6) as repeated fixed factor, and Age, Sex and PTA as covariates.

- Results. Main effect of Span for Acc ($F3, 110.8 = 189.32$, $p \leq .001$) (Figure 2a) and RT ($F3, 88.4 = 113.323$, $p \leq .001$) (Figure 2b), with a decline in performance with an increase in Span.
- Main effect of Age for Acc ($F1, 71.5 = 9.311$, $p = .003$, $\beta = -.002$) (Figure 2c) and RT ($F1, 74.8 = 18.602$, $p \leq .001$, $\beta = 61.525$) (Figure 2d), with a decline in performance with age.

Figure 2 : Behavioural Results

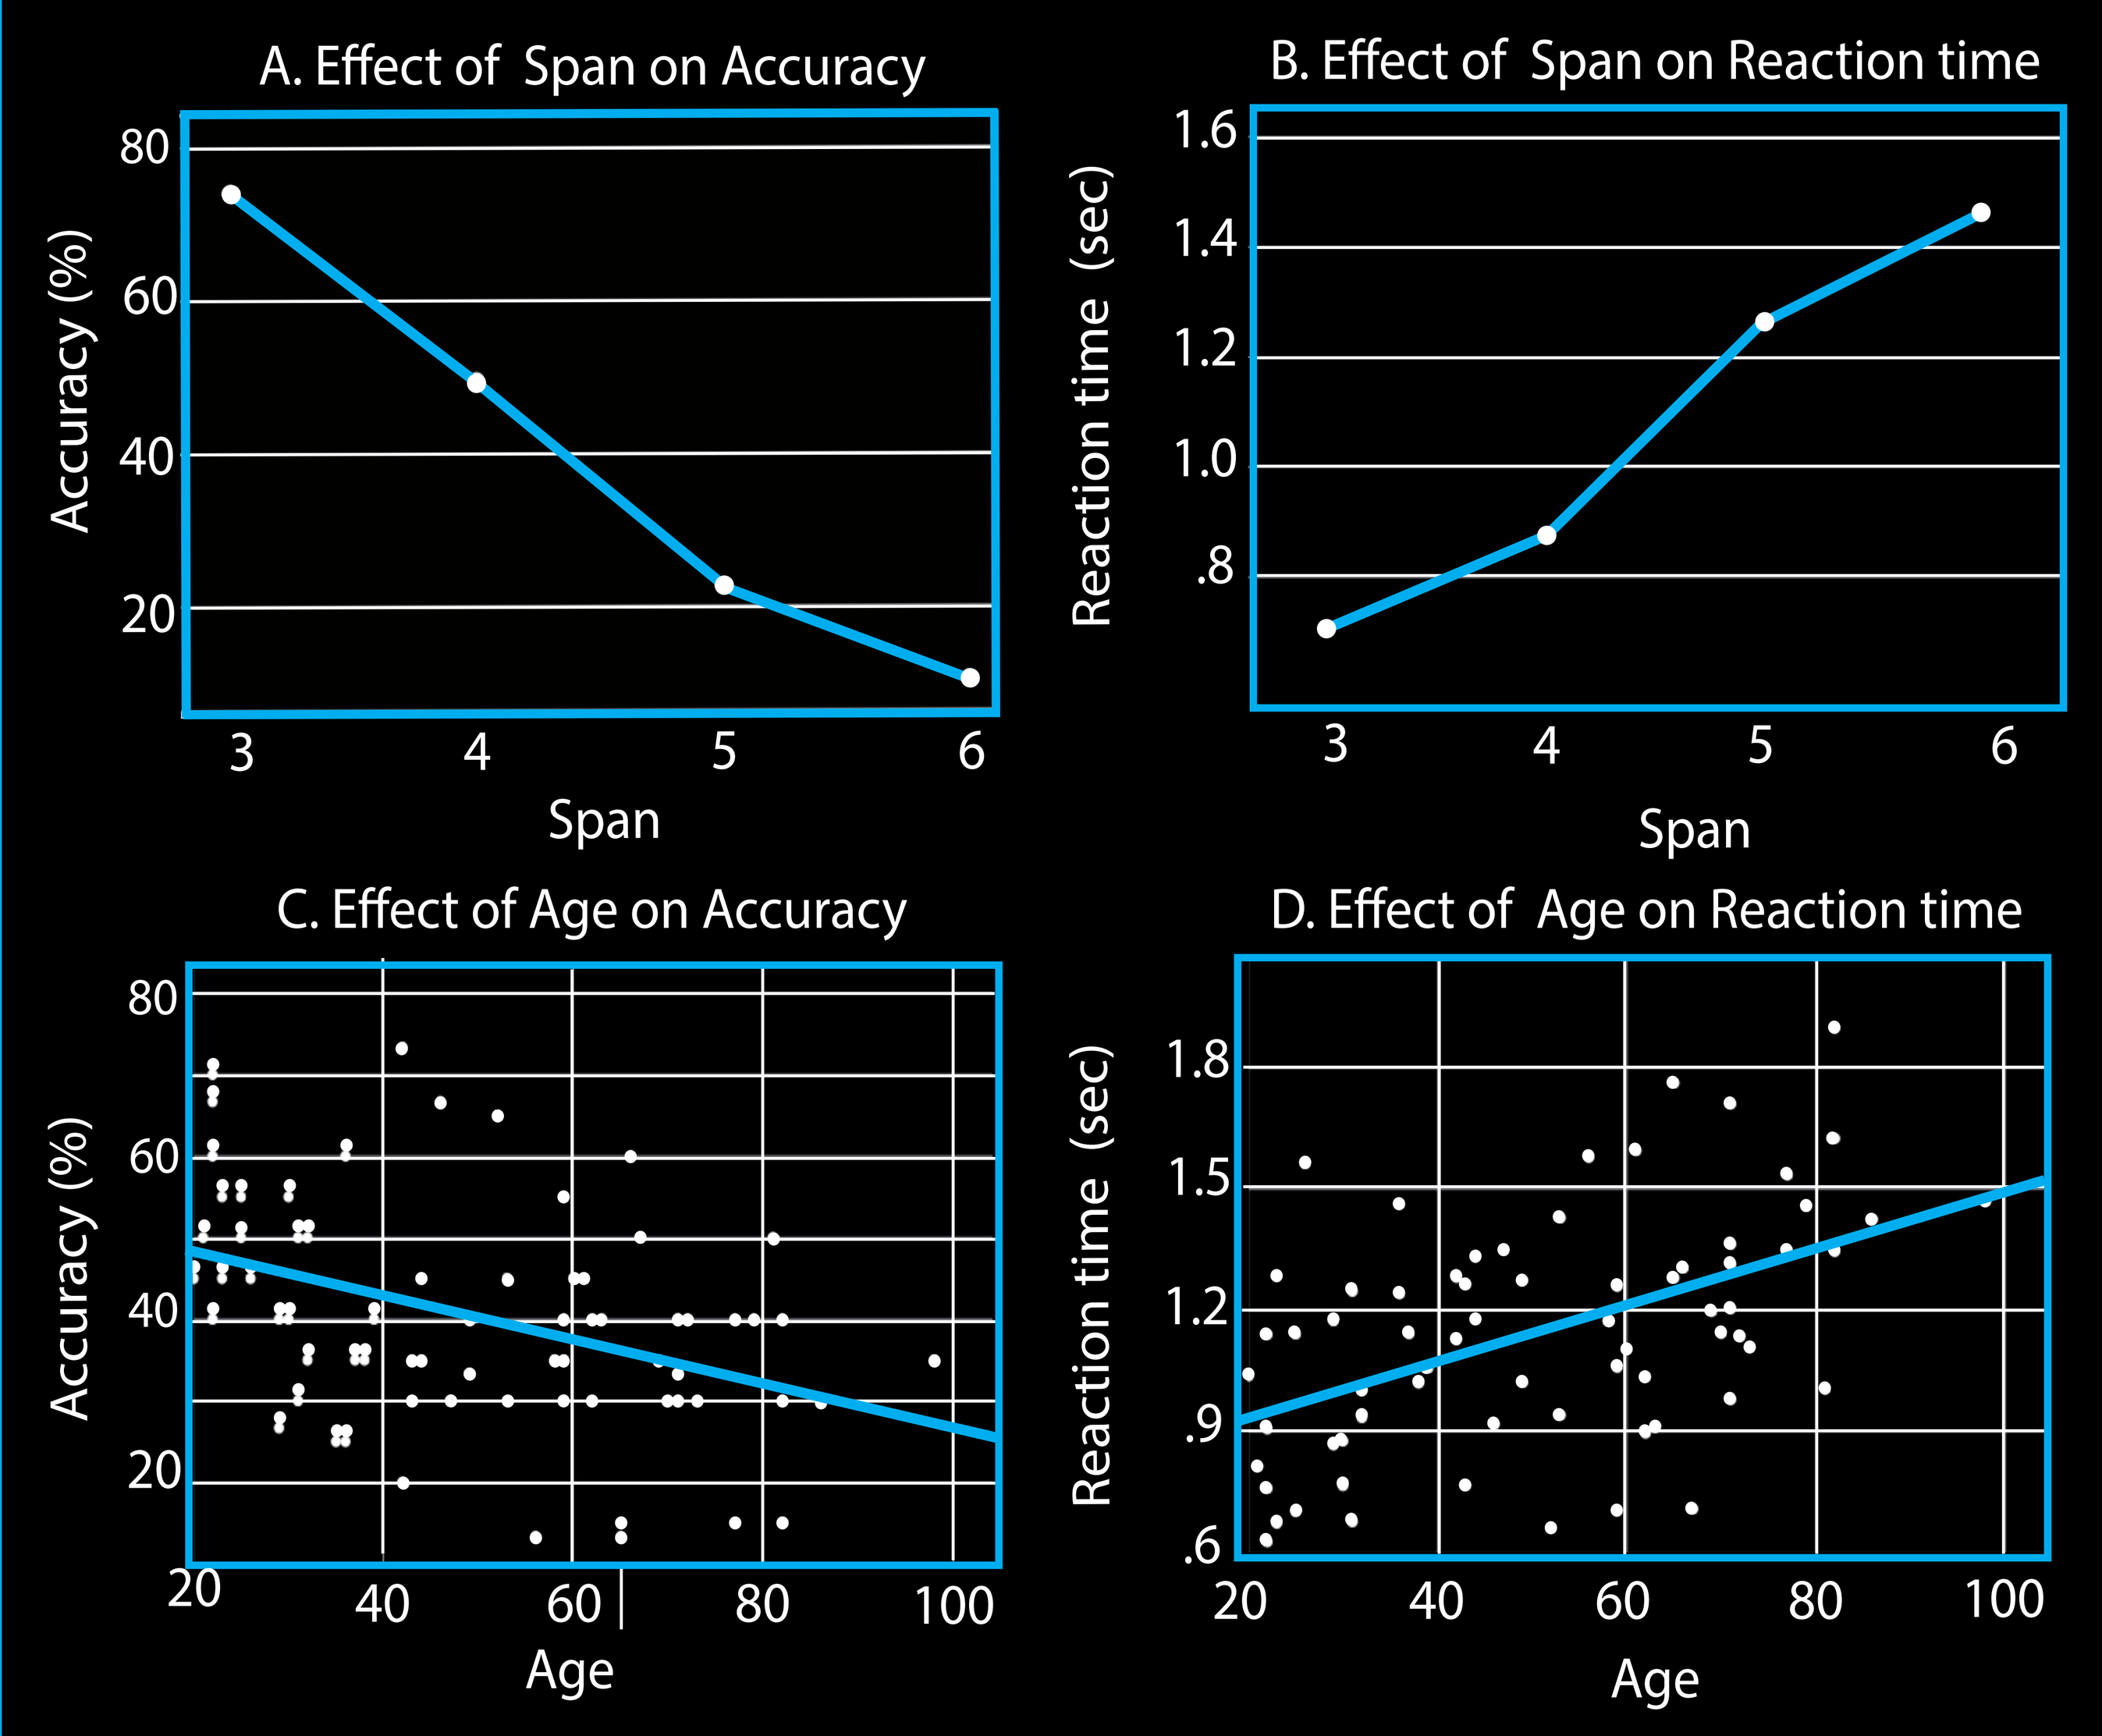
A. Effect of Span on Accuracy
Accuracy (%)
Span
B. Effect of Span on Reaction time
Reaction time (sec)
Span
C. Effect of Age on Accuracy
Accuracy (%)
Age
D. Effect of Age on Reaction time
Reaction time (sec)
Age

Study 2 (MRI)

- Moderation analysis (Figure 3) to determine if cortical aging affects avWM.

- Results (Table 1, Figure 4). The relationship between age and avWM varies as a function of the structure (Surface, Volume, Thickness) in several regions including bilateral middle frontal gyrus (MFG) (Figure 5a, 5b), left angular gyrus (AnG) (Figure 5c), left anterior cingulate cortex (ACC) (Figure 5d) and bilateral temporal transverse gryrus (TTG) (Figure 5e, 5f).

- In some regions, age-related decline in avWM was less intense in those with higher volume/surface (bilateral MFG). In other regions, stronger decline in performance was found in those with higher volume/thickness (left AnG, left Acc, bilateral TTG).

Figure 3 : Moderation model

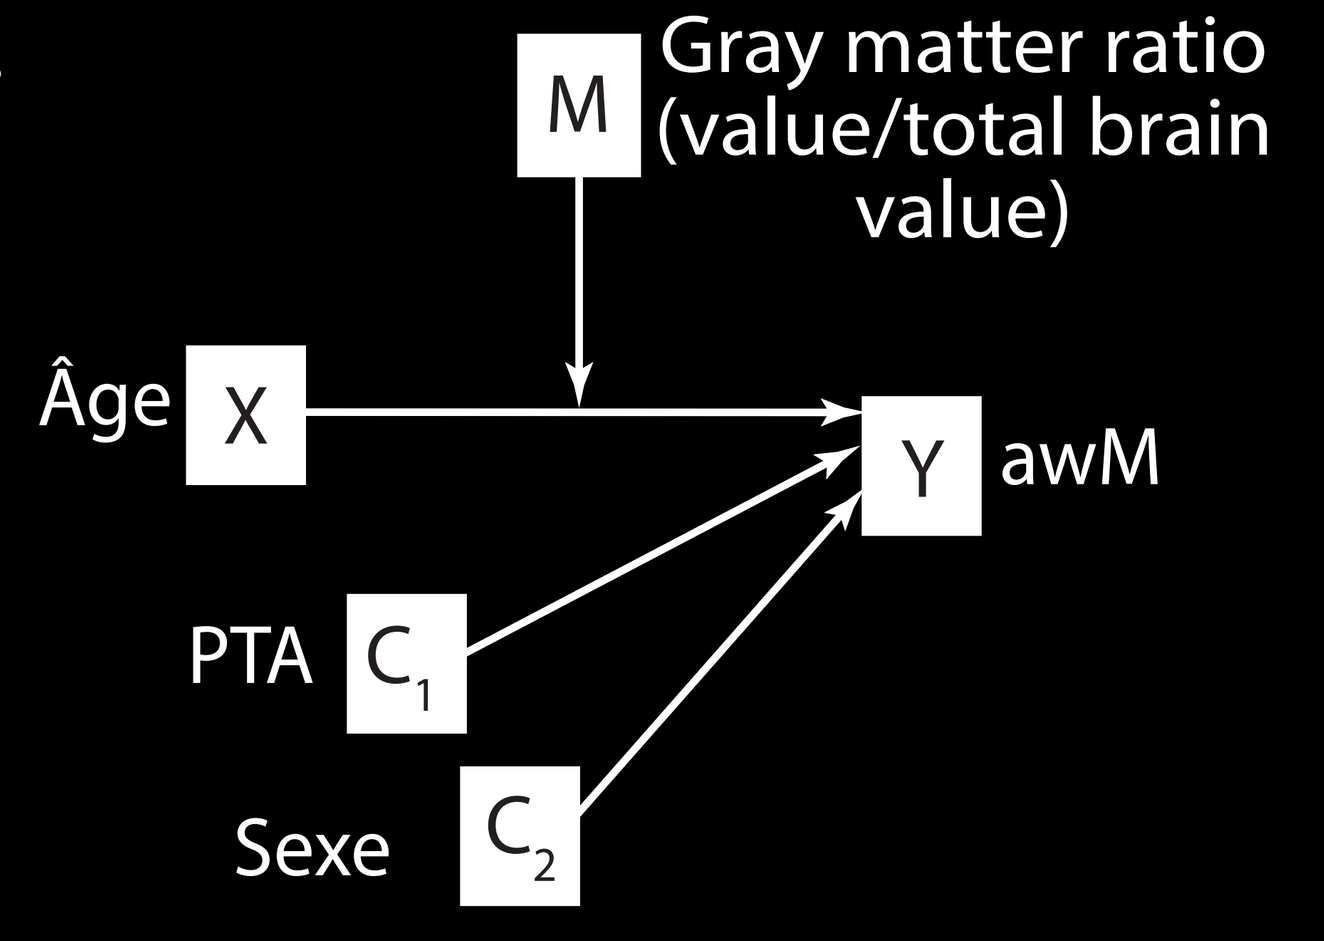
M
Gray matter ratio (value/total brain value)
Âge
X
Y
awM
PTA
C_1
Sexe
C_2

Figure 4 : Neurobiological correlates

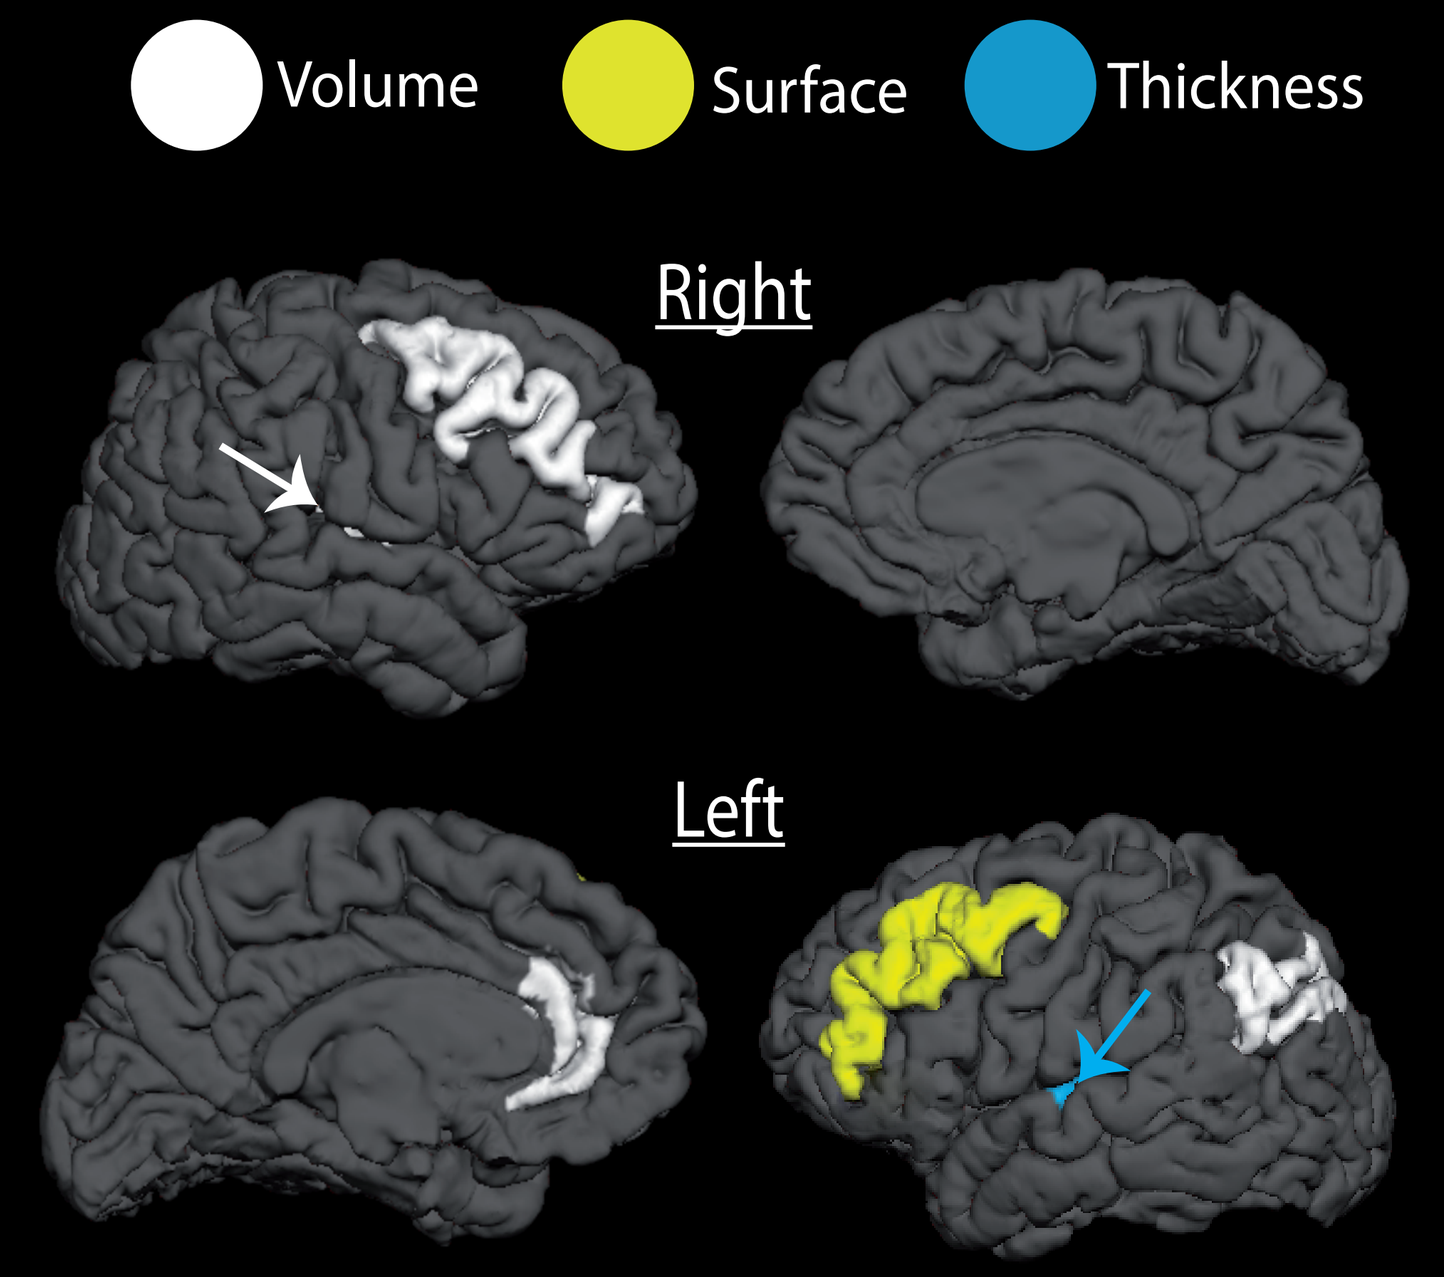
Volume
Surface
Thickness
Right
Left

Figure 5 : Moderation results

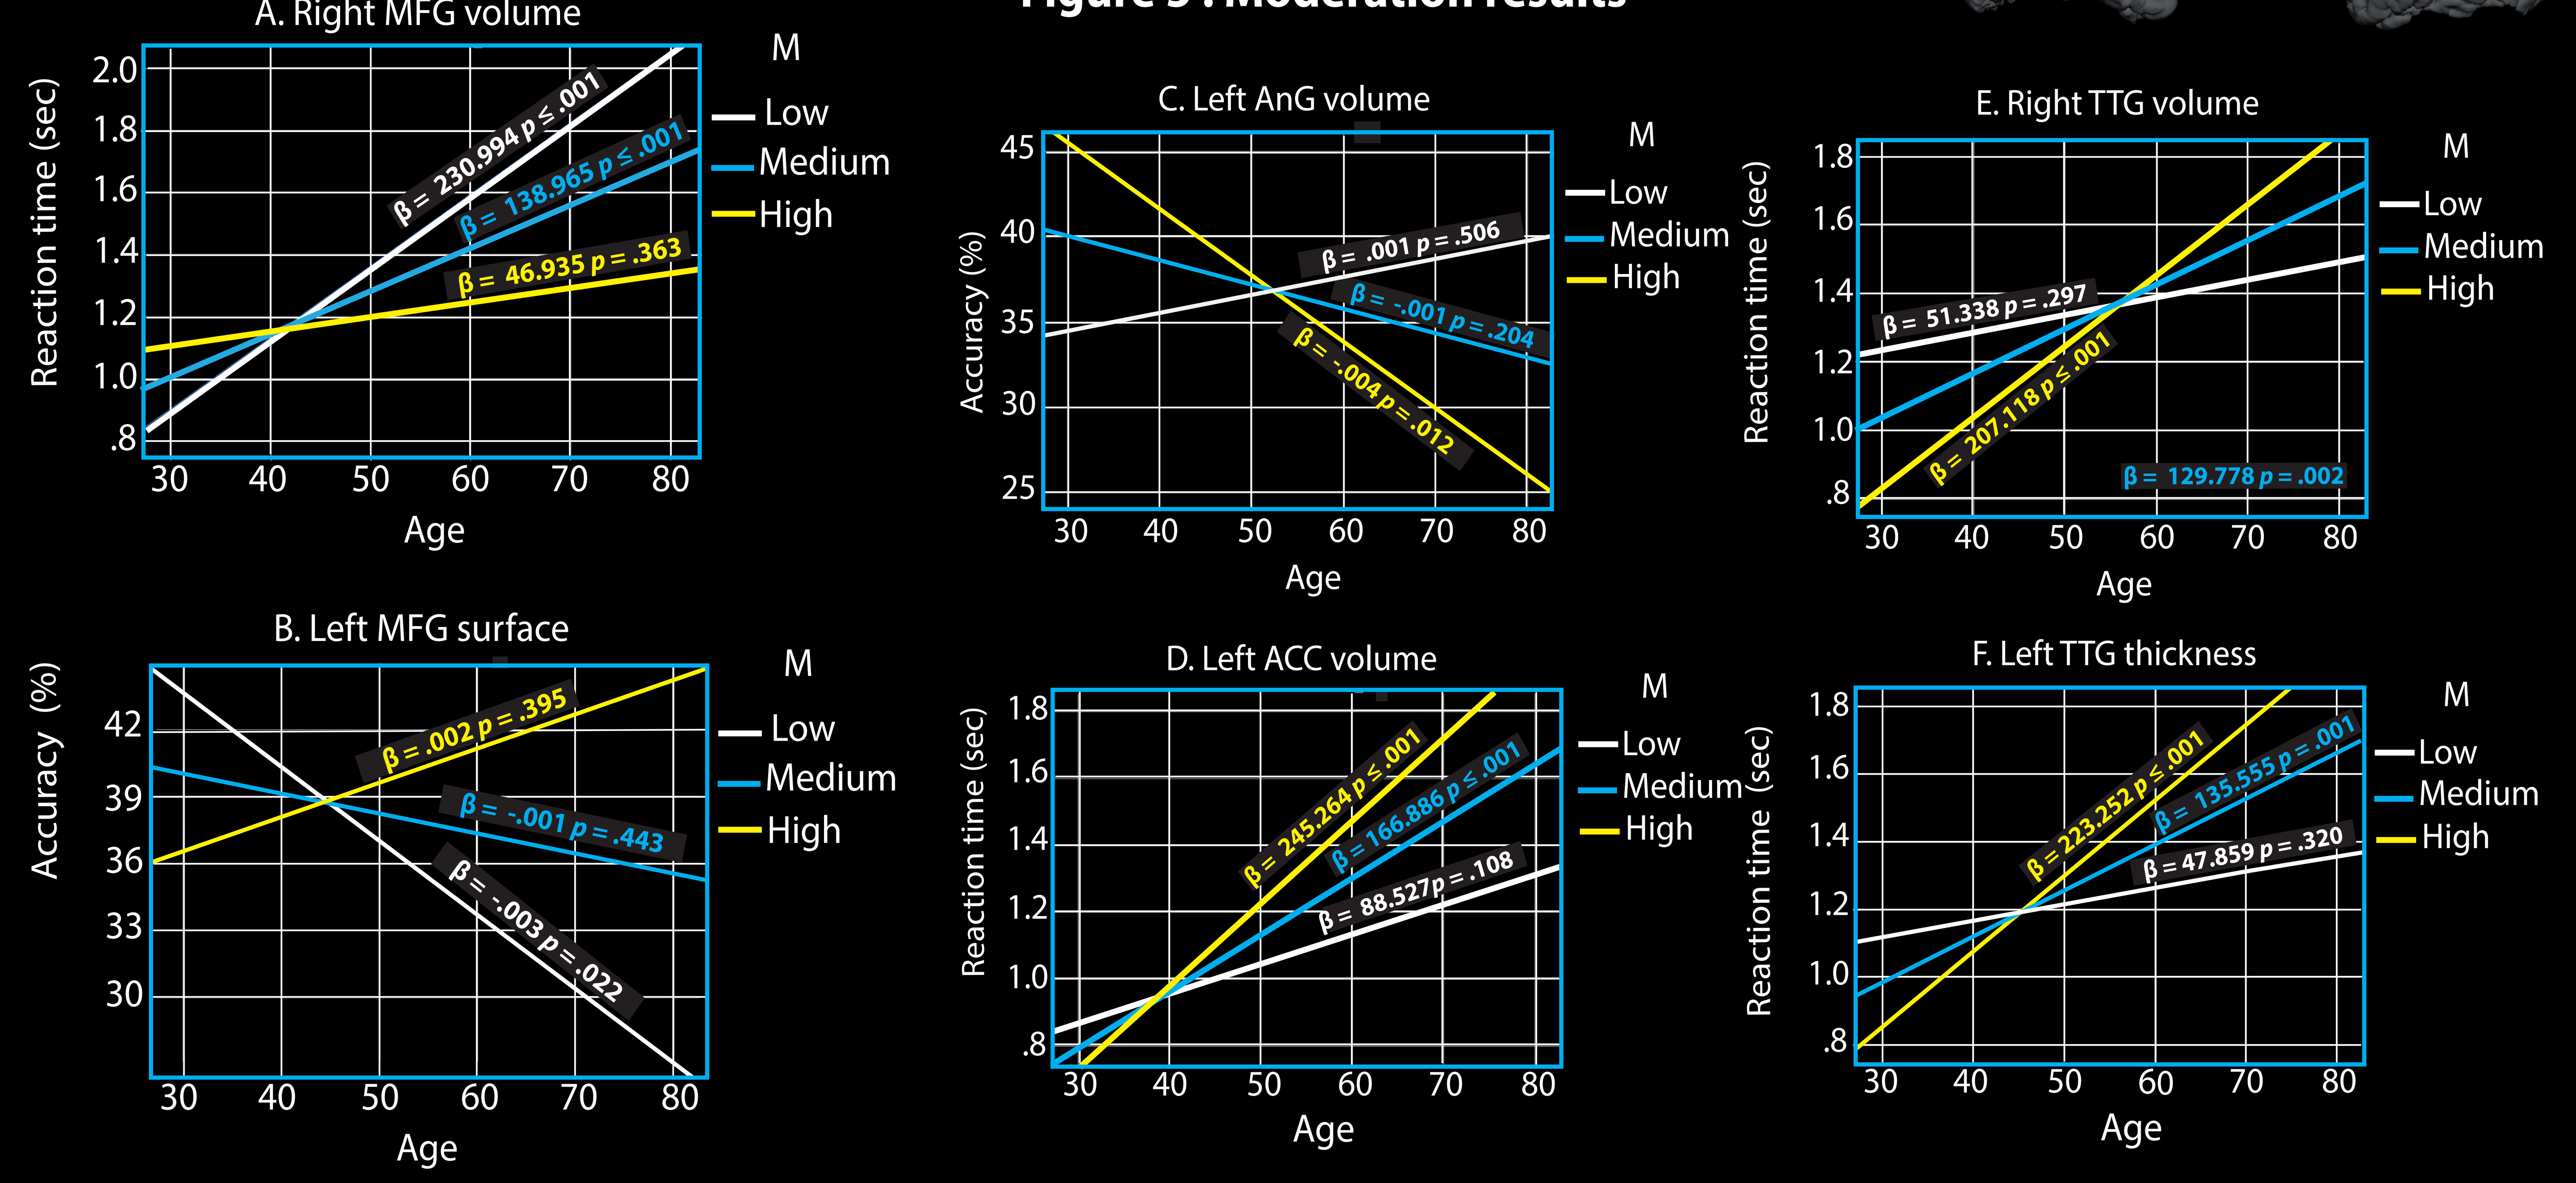
A. Right MFG volume
Reaction time (sec)
Age
M
Low
Medium
High
β = 230.994 p ≤ .001
β = 138.965 p ≤ .001
β = 46.935 p = .363
B. Left MFG surface
Accuracy (%)
β = .002 p = .395
β = -.001 p = .443
β = -.003 p = .022
C. Left AnG volume
Accuracy (%)
β = .001 p = .506
β = -.001 p = .204
β = -.004 p = .012
D. Left ACC volume
Reaction time (sec)
β = 245.264 p ≤ .001
β = 166.886 p ≤ .001
β = 88.527p = .108
E. Right TTG volume
Reaction time (sec)
β = 51.338 p = .297
β = 207.118 p ≤ .001
β = 129.778 p = .002
F. Left TTG thickness
Reaction time (sec)
β = 223.252 p ≤ .001
β = 135.555 p = .001
β = 47.859 p = .320

Conclusion

- Normal aging is associated with a decline in accuracy and reaction time in avWM.
- This decline is moderated by cortical structure in different ways, suggesting that the impact of brain aging on cognitive functions is complex and spatially heterogeneous.
- We found a positive impact of the structure of the bilateral MFG on avWM, consistent with the HAROLD model.
- The MFG is involved in manipulating and monitoring verbal information in avWM [8, 9].
- Negative impacts for the other regions were found, which are under investigation.

References

[1] Hedden T., et al. (2001). Psychology and Aging. [2] Cabeza, R. (2002). Psychology and Aging. [3] Burton, MW., et al. (2005) NeuroImage. [4] Deschamps, I., et al. (2014) Neuropsychologia. [5] Nasredinne, Z. S., et al. (2005) Journal of the American Geriatrics Society. [6] Pollack, I. et al. (1959). Journal of Experimental Psychology. [7] Destrieux, C. et al. (2009). NeuroImage. [8] Petrides et al. (1993). Proceedings of the National Academy of Sciences. [9] Barbey et al. (2013). Cortex.

Acknowledgments

FONDATION DRUMMOND FOUNDATION
INNOVATION.CA CANADA FOUNDATION FOR INNOVATION | FONDATION CANADIENNE POUR L'INNOVATION
Fonds de recherche Santé Québec
CRSNG NSERC